EFFICACY OF QUADRIVALENT HUMAN PAPILLOMAVIRUS VACCINE AMONG KENYAN ADOLESCENTS AND YOUNG ADULTS LIVING WITH HIV NINE YEARS AFTER INITIAL VACCINATION

Lynda M. Oluoch[1] | Torin T. Schaafsma[2] | Katherine K. Thomas[2] | Denise A. Galloway[3] | Paul Mwangi[1] | Kenneth Ngure[2,4] | Jane N. Gacheru[1] | Irene Njeru[1] | Jacinta Nyokabi[1] | Jason Caucutt[2] | Marianne Mureithi[2,5] | Linda Eckert[2,6] | Ruanne V. Barnabas [7,8] | Anna Wald[9,10,11,12] | Nelly R. Mugo[1,2]

[1]Centre of Clinical Research, Kenya Medical Research Institute (KEMRI), Nairobi, Kenya, [2]Department of Global Health, University of Washington, Seattle, WA, [3]Fred Hutchinson Cancer Center, University of Washington, Seattle, WA, [4]School of Public Health Jomo Kenyatta University of Agriculture and Technology, Nairobi, Kenya, [5]Department of Medical Microbiology & Immunology, University of Nairobi, Kenya, [6]Departments of Obstetrics and Gynaecology, University of Washington, Seattle, WA, [7]Division of Infectious Diseases, Massachusetts General Hospital, Boston, [8]Harvard Medical School, Boston, [9]Department of Medicine, University of Washington, Seattle, WA, [10]Department of Laboratory Medicine and Pathology, University of Washington, Seattle, WA, [11]Department of Epidemiology, University of Washington, Seattle, WA, [12]Vaccine and Infectious Diseases Division, Fred Hutchinson Cancer Research Center, Seattle, WA

BACKGROUND

- People living with HIV (PLWHIV) experience a high burden of HPV-associated disease.
- Primary prevention with the HPV vaccine is recommended for preadolescents prior to sexual debut, offering long-term protection against vaccine-targeted HPV genotypes.
- However, data on the durability of protection in PLWHIV is limited.
- In 2014, we enrolled 180 adolescents living with HIV (aged 9–14) who received three doses of the quadrivalent HPV vaccine.
- This cohort was invited for re-enrolment nine years post-vaccination to assess sustained protection.

METHODS

- In 2023, 158 adolescents and young adults willing to continue follow-up were re-enrolled.
- Genital swabs were collected from 155 participants and assessed for HPV DNA by PCR at three timepoints: enrollment, month six, and month twelve; alongside blood for HIV viral load, CD4 count and HPV antibodies at corresponding intervals.
- We evaluated 28 HPV genotypes (Fig 1), with a focus on the four vaccine-specific HPV genotypes in the administered vaccine (6, 11, 16, 18).
- Presence of persistent HPV infection was determined as detection of the same HPV genotype at consecutive visits at least six months apart.

RESULTS

- Median age at re-enrolment was 20 years IQR [19–22]; >99% were on antiretroviral treatment and 65% had plasma HIV RNA < 40 copies/mL, with a median CD4 cell count of 687 [IQR 432–930].
- Two participants (both women) had vaccine genotype persistent infections, both of which were in low-risk genotype HPV-6; with one having high HIV RNA (>10,000 copies/ml) and the other with viral suppression during the 1-year follow-up.

Table 1. Baseline Characteristics

	Women (n=84)	Men (n=74)
Age	20 (19–22)	20 (19–22)
Ever sexually active (self-report)	58 (69%)	38 (51%)
Currently on ART	84 (100%)	73 (99%)
Years on ART (among those currently on ART)	13.0 (10.4–14.8)	13.4 (12.0–15.6)
HIV viral load <40 copies/mL	49 (58%)	54 (73%)
CD4 count	772 (419–1046)	619 (444–818)
HSV2 positive	11/78 (14%)	1/72 (1.4%)
Chlamydia positive	7/83 (8.4%)	0 (0%)
Gonorrhea positive	5/83 (6.0%)	0 (0%)

statistics are n and (%) or median and (interquartile range), denominator provided if any missing

Out of 151 participants, only 2 participant had vaccine genotype persistent infections, both of which were in low-risk genotype HPV-6

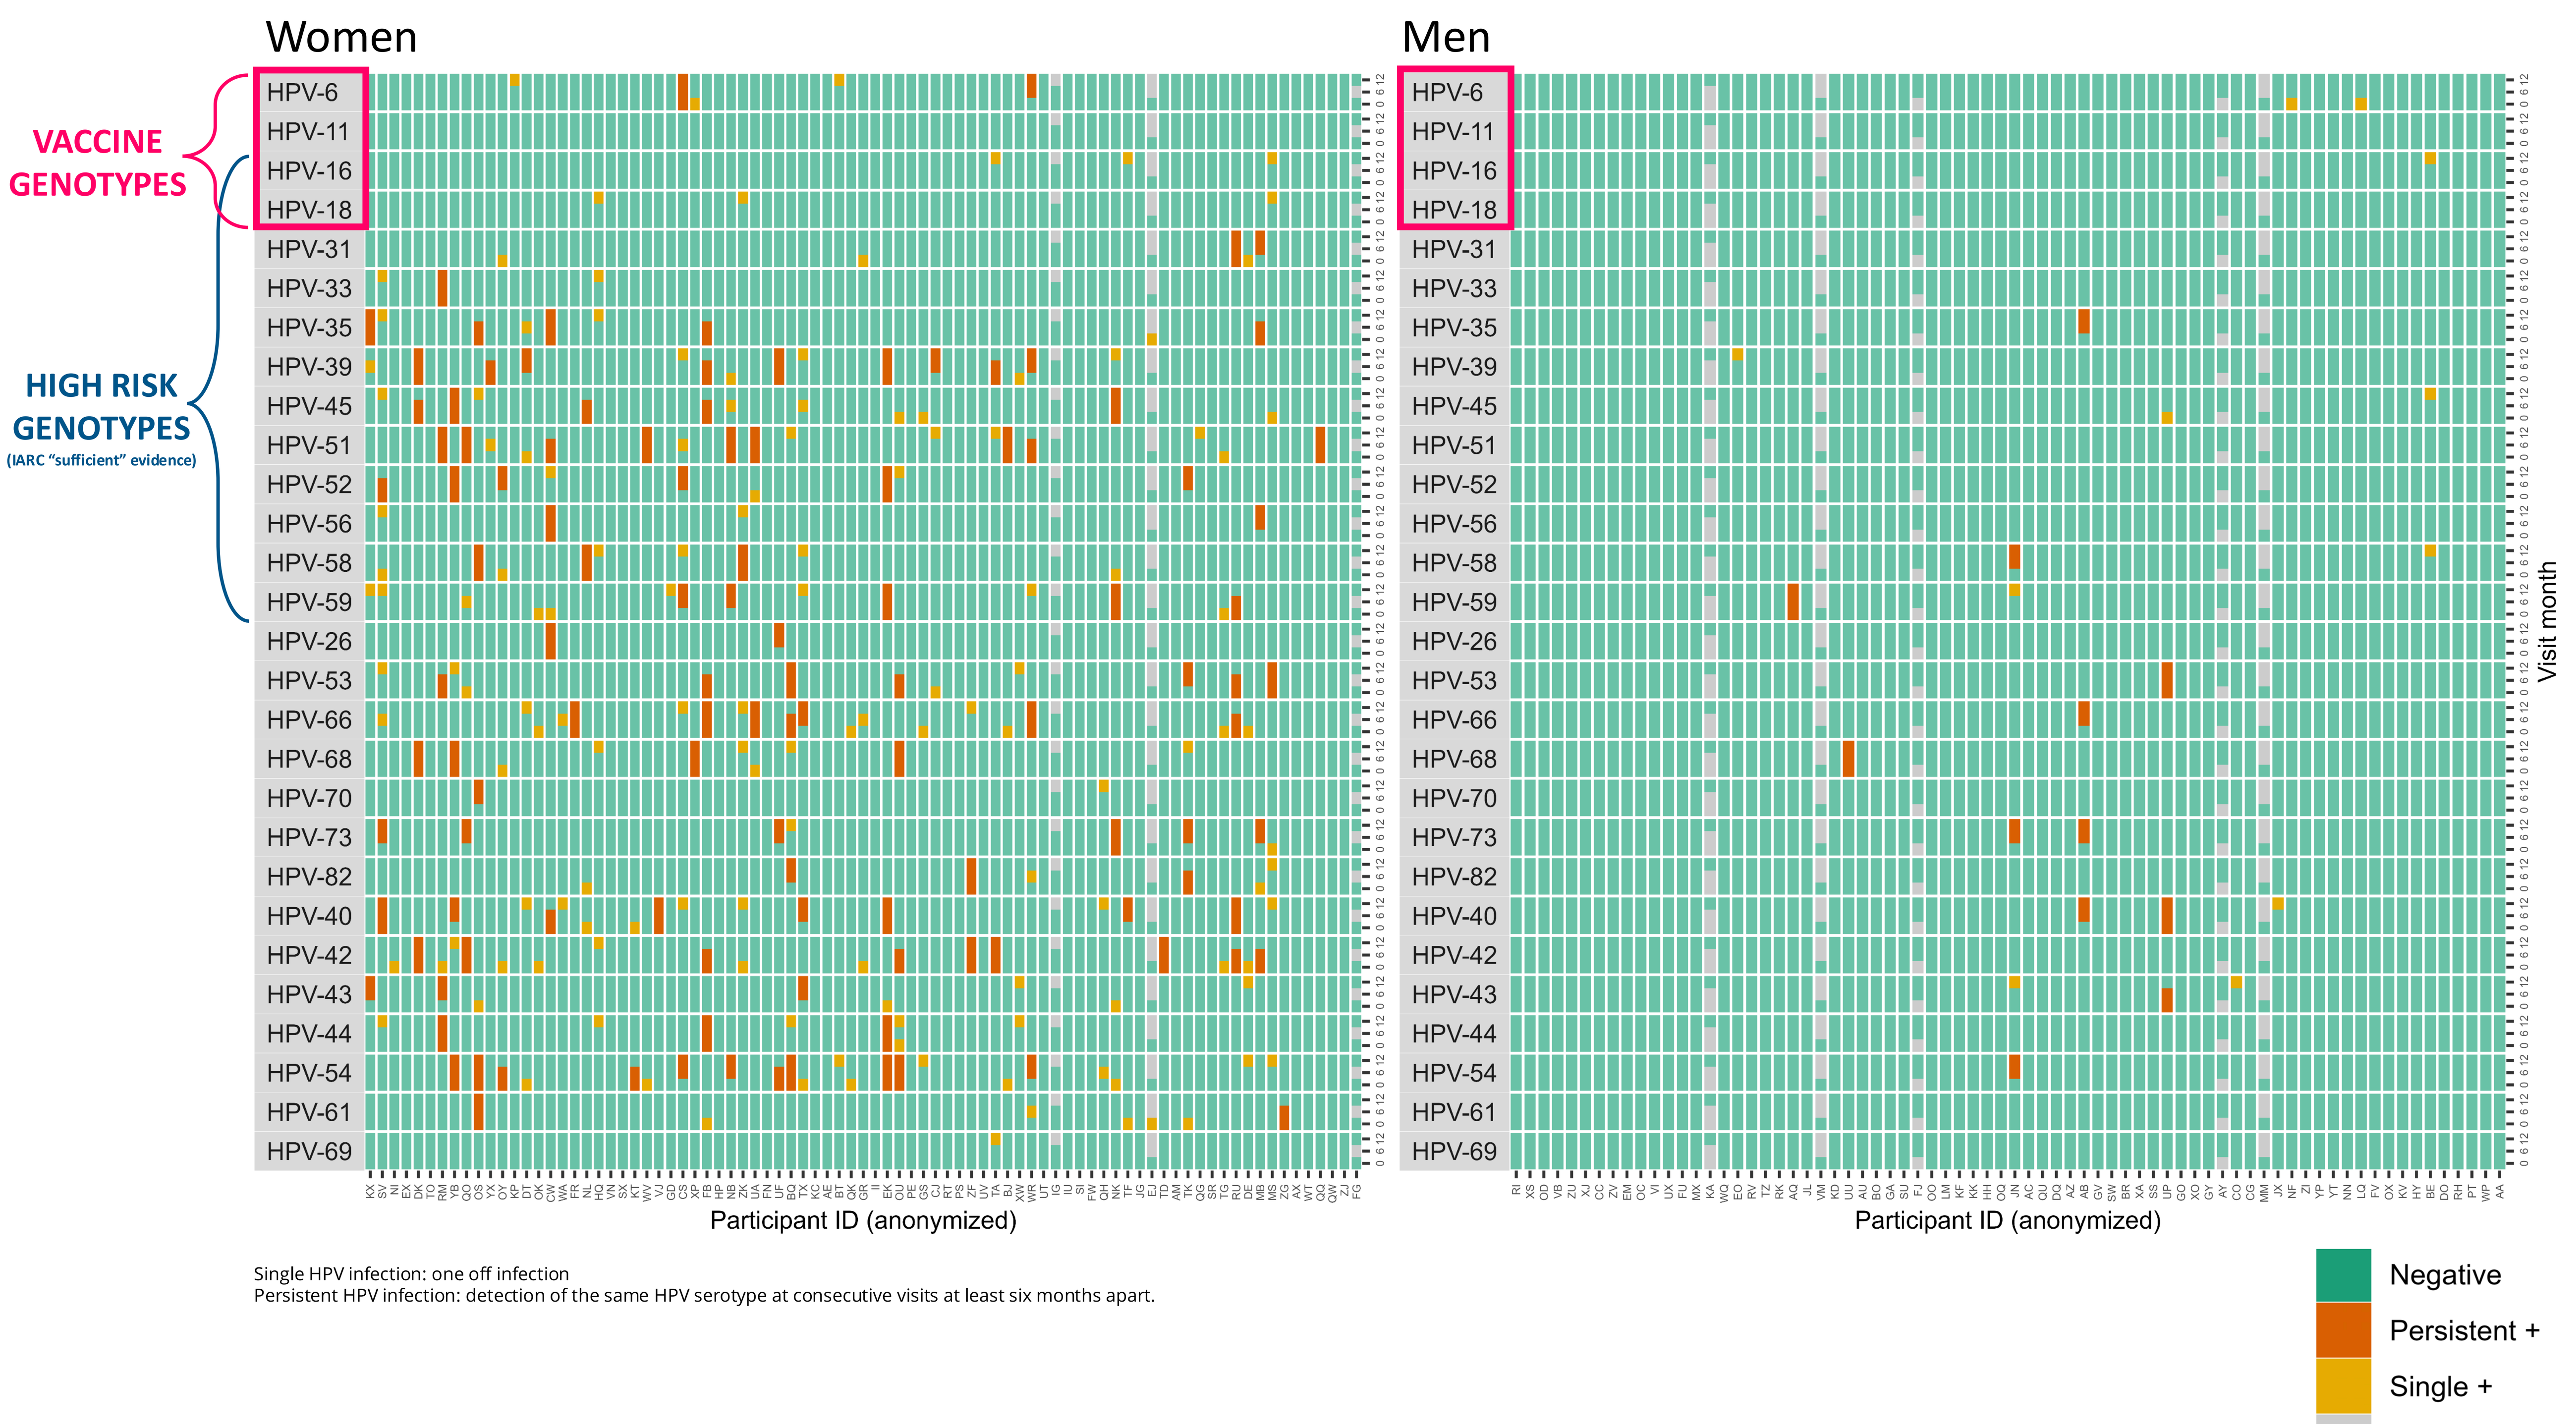
Women
Men
VACCINE GENOTYPES
HIGH RISK GENOTYPES
(IARC "sufficient" evidence)
HPV-6
HPV-11
HPV-16
HPV-18
HPV-31
HPV-33
HPV-35
HPV-39
HPV-45
HPV-51
HPV-52
HPV-56
HPV-58
HPV-59
HPV-26
HPV-53
HPV-66
HPV-68
HPV-70
HPV-73
HPV-82
HPV-40
HPV-42
HPV-43
HPV-44
HPV-54
HPV-61
HPV-69
Participant ID (anonymized)
Visit month
Single HPV infection: one off infection
Persistent HPV infection: detection of the same HPV serotype at consecutive visits at least six months apart.
Negative
Persistent +
Single +
[No data]

Figure 1: Single and persistent HPV infection by genotype

Table 2. Prevalence of single and persistent infection by sex and HPV vaccine genotypes

	Total (N enrolled = 158)		Women (n enrolled = 84)		Men (n enrolled = 74)	
	n/N	Prev (95% CI)	n/N	Prev (95% CI)	n/N	Prev (95% CI)
Persistent HPV infection (2+ consecutive positives 6+ months apart)						
HPV-6	2/151	1.3% (0.2–5.2)	2/82	2.4% (0.4–9.4)	0/69	0.0% (0.0–6.6)
HPV-11	0/151	0.0% (0.0–3.1)	0/82	0.0% (0.0–5.6)	0/69	0.0% (0.0–6.6)
HPV-16	0/151	0.0% (0.0–3.1)	0/82	0.0% (0.0–5.6)	0/69	0.0% (0.0–6.6)
HPV-18	0/151	0.0% (0.0–3.1)	0/82	0.0% (0.0–5.6)	0/69	0.0% (0.0–6.6)
Any vaccine genotype	2/151	1.3% (0.2–5.2)	2/82	2.4% (0.4–9.4)	0/69	0.0% (0.0–6.6)
Any non-vaccine genotype	46/151	30.5% (23.4–38.6)	41/82	50.0% (39.4–60.6)	5/69	7.2% (2.7–16.8)
Any HPV infection						
HPV-6	7/155	4.5% (2.0–9.4)	5/83	6.0% (2.2–14.1)	2/72	2.8% (0.5–10.6)
HPV-11	0/155	0.0% (0.0–3.0)	0/83	0.0% (0.0–5.5)	0/72	0.0% (0.0–6.3)
HPV-16	4/155	2.6% (0.8–6.9)	3/83	3.6% (0.9–10.9)	1/72	1.4% (0.1–8.5)
HPV-18	3/155	1.9% (0.5–6.0)	3/83	3.6% (0.9–10.9)	0/72	0.0% (0.0–6.3)
Any vaccine genotype	13/155	8.4% (4.7–14.2)	10/83	12.0% (6.2–21.5)	3/72	4.2% (1.1–12.5)
Any non-vaccine genotype	65/155	41.9% (34.1–50.1)	56/83	67.5% (56.2–77.1)	9/72	12.5% (6.2–22.9)

For persistent HPV infection: N = Participants with at least two timepoints
For any HPV infection: N = Participants with at least one timepoint
Prev = Prevalence
95% CI = Wilson 95% confidence intervals

CONCLUSION:

Among adolescent girls and boys living with HIV, three doses of the HPV vaccine offered sustained protection nine years post-vaccination against vaccine-specific high-risk HPV infections. These findings highlight the importance of HPV vaccination programs targeting adolescents living with HIV in similar contexts, reinforcing the potential for long-term disease prevention in this high-risk population.

REFERENCES

- World Health Organization. (2023)
- Staadegaard, Lisa, et al, EClinicalMedicine 2022
- Mugo Nelly et al, Vaccine 2021

ACKNOWLEDGEMENTS

- **Study participants**
- **MISP Funding #60583**
- **KEMRI-PHRD Study Staff**

Supported in part by a research grant from Investigator-Initiated Studies Program of Merck Sharp & Dohme LLC. The opinions expressed in this paper are those of the authors and do not necessarily represent those of Merck Sharp & Dohme LLC.

Contact: lynda@pipsthika.org

Efficacité durable du vaccin quadrivalent contre le HPV chez des adolescents kényans vivant avec le VIH

Auteurs : Lynda M. Oluoch, Torin T. Schaafsma, Katherine K. Thomas, Denise A. Galloway, Paul Mwangi, Kenneth Ngure, Jan Cheru, Irene Njeru, Jacinta Nyokabi, Jason Caucutt, Marianne Mureithi, Linda Eckert, Ruanne V. Barnabas, Abigail Wald, Nelly R. Mugo

Affiliations : Centre de recherche clinique, Kenya Medical Research Institute (KEMRI), Nairobi ; Département de santé mondiale, University of Washington (Seattle, États-Unis) ; Fred Hutchinson Cancer Center, University of Washington (Seattle, États-Unis) ; School of Public Health de l'Université Jomo Kenyatta d'agriculture et de technologie, Nairobi ; Département de microbiologie et d'immunologie médicales de l'Université de Nairobi ; Départements d'obstétrique et de gynécologie et de médecine de l'Université de Washington (Seattle) ; Division des maladies infectieuses du Massachusetts General Hospital et Harvard Medical School (Boston, États-Unis) ; Vaccine and Infectious Diseases Division, Fred Hutchinson Cancer Research Center (Seattle).

Résumé détaillé :

Le papillomavirus humain (HPV) provoque des lésions précancéreuses et des cancers, et les adolescents vivant avec le VIH ont un risque particulièrement élevé. Dans cette cohorte kényane, 180 adolescents de 9 à 14 ans vivant avec le VIH ont reçu trois doses du vaccin quadrivalent (types 6, 11, 16 et 18) et ont été invités à un suivi neuf ans plus tard. Les auteurs ont réévalué 158 participants en prélevant des écouvillons cervicaux et en mesurant l'ADN du HPV et les anticorps. Les infections persistantes étaient définies comme la détection d'un même génotype à deux visites espacées d'au moins six mois. Les résultats montrent que moins de 2 % des participants présentaient une infection persistante par le HPV 6 et qu'aucune infection persistante par les types 11, 16 ou 18 n'a été détectée. Globalement, seulement 1,3 % des participants présentaient une infection par un génotype contenu dans le vaccin. Ces chiffres indiquent une protection durable contre les génotypes du vaccin neuf ans après la vaccination. La quasi-totalité des participants suivaient un traitement antirétroviral depuis une douzaine d'années et la majorité avaient une charge virale indétectable. Les auteurs concluent que, même dans un contexte de VIH, trois doses du vaccin quadrivalent offrent une protection à long terme contre les génotypes du vaccin et justifient la poursuite des programmes de vaccination ciblant les adolescents vivant avec le VIH.

Cette traduction a été réalisée avec l'aide d'une IA. Merci de signaler toute inexactitude aux organisateurs afin que nous puissions la corriger rapidement, ou à l'adresse suivante : CHIC-SPC.secretariat@uantwerpen.be.